TOTUM-63 DIFFERENTIALLY ALTERS INCRETIN AND HORMONAL RESPONSES TO GLUCOSE STIMULATION IN HIGH FAT-FED MICE

B. Morio[1], V. Chavanelle[2], M. Soumarmon[1], S. Peltier[2], P. Sirvent[2]

[1]CarMeN Laboratory, INSERM U1060, INRA U1397, Lyon 1 University, France; [2]Valbiotis, Périgny, France.

CONTEXT

TOTUM-63 has been demonstrated to significantly improve body weight and glucose homeostasis in animal models of obesity and type 2 diabetes (db/db and C57BL/6 fed a high fat diet). Our study aimed at exploring whether the mechanisms include improved hormone secretion by the digestive system. Specifically, incretins (glucose-dependent insulinotropic peptide (GIP), glucagon-like petide-1 (GLP-1)) and peptide hormones (cholecystokinin (CCK) and peptide YY (PYY)) have been shown to be major regulators of glucose control.

MATERIAL AND METHODS

Thirty male C57BL/6JRJ mice, aged 6 weeks on the start day, were assigned to standard chow (CON, n=10), high fat diet (HF, 60% energy from fat, n=10) or HF-TOTUM-63 (HF diet 60% supplemented with TOTUM-63 2.7%, n=10) groups for 10 weeks (ethical agreement CECCAP LS_2017_004).

At the end of the dietary intervention, glucose homeostasis was assessed during an oral glucose tolerance test (OGTT) and an intraperitoneal pyruvate tolerance test (ipPTT). Doses of glucose and pyruvate were adjusted to the lean body mass, so that when expressed to body weight, CON and HF-TOTUM-63 received 2g/kg and HF 1.7g/kg of glucose or pyruvate. During the OGTT, peripheral insulin concentration was assessed before and 15 and 120 min after glucose gavage on blood samples taken at the tail in the presence of EDTA.

Thereafter, overnight fasted mice received an oral gavage of glucose (2g/kg body weight) or vehicle (water) 15 min before being anesthetized. Blood from the portal vein was drawn in the presence of EDTA and DPP-IV inhibitor for assessment of GIP, GLP-1, CCK, PYY, insulin and glucagon concentrations.

Statistical analyses: Data are presented as means ± SEM. One-way or two-way (when appropriate) analyses of variance (ANOVA) were performed to test the effect of the experimental conditions. When a significant effect was detected, a posteriori Bonferroni correction test was used to analyze pairwise differences. For other comparisons, data were analysed with Student t test. Statistical analysis was performed using GraphPad Prism® software (USA). P values<0.05 were considered significant.

CONCLUSION

As expected TOTUM-63 improved glucose homeostasis of high-fat fed animals. This was associated with the restauration to CON values of GLP-1, insulin and glucagon circulating concentration in the portal vein. However, incretin and hormonal responses were differentially affected as GIP, CCK and PYY responses to glucose challenge differed between HF-TOTUM-63 and CON.

RESULTS

1. Body weight

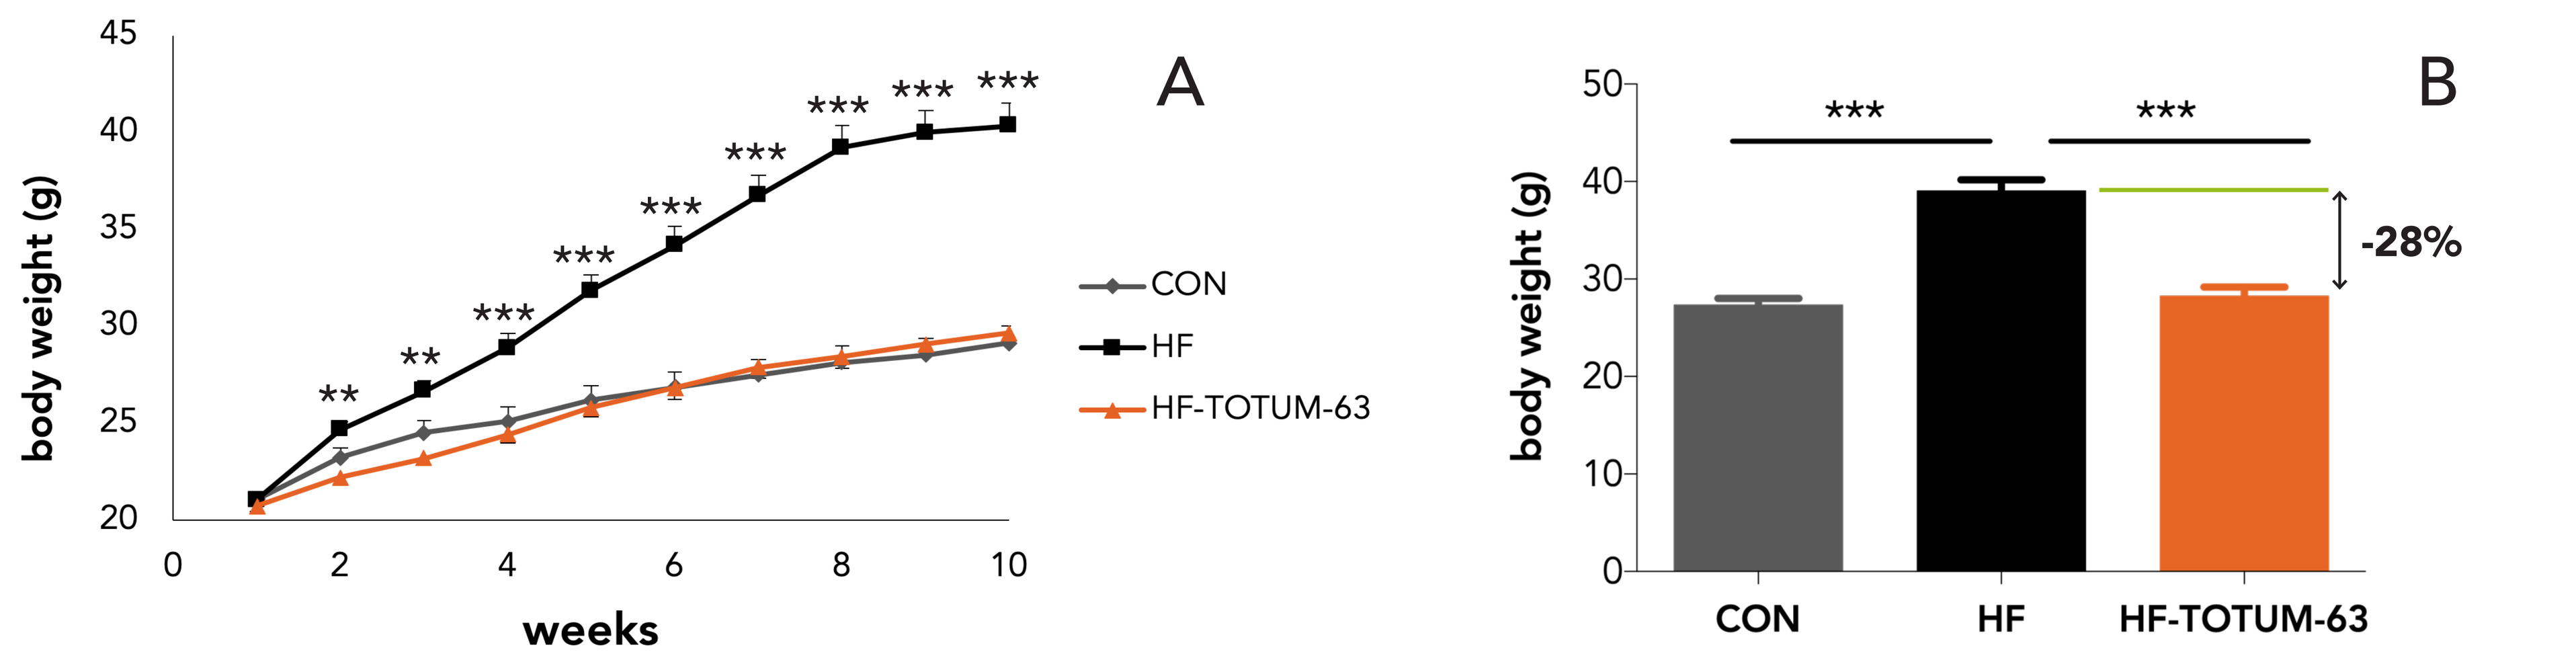
A
body weight (g)
weeks
CON
HF
HF-TOTUM-63
B
body weight (g)
-28%
CON
HF
HF-TOTUM-63

Fig. 1 – Impact of TOTUM-63 on (A) the kinetics of body weight gain and (B) the body weight at the end of the experimentation.

***** : p< 0.001**

The body weight of mice fed HF-TOTUM-63 was similar to that of CON mice and 28% lower than that of HF.

2. Food intake

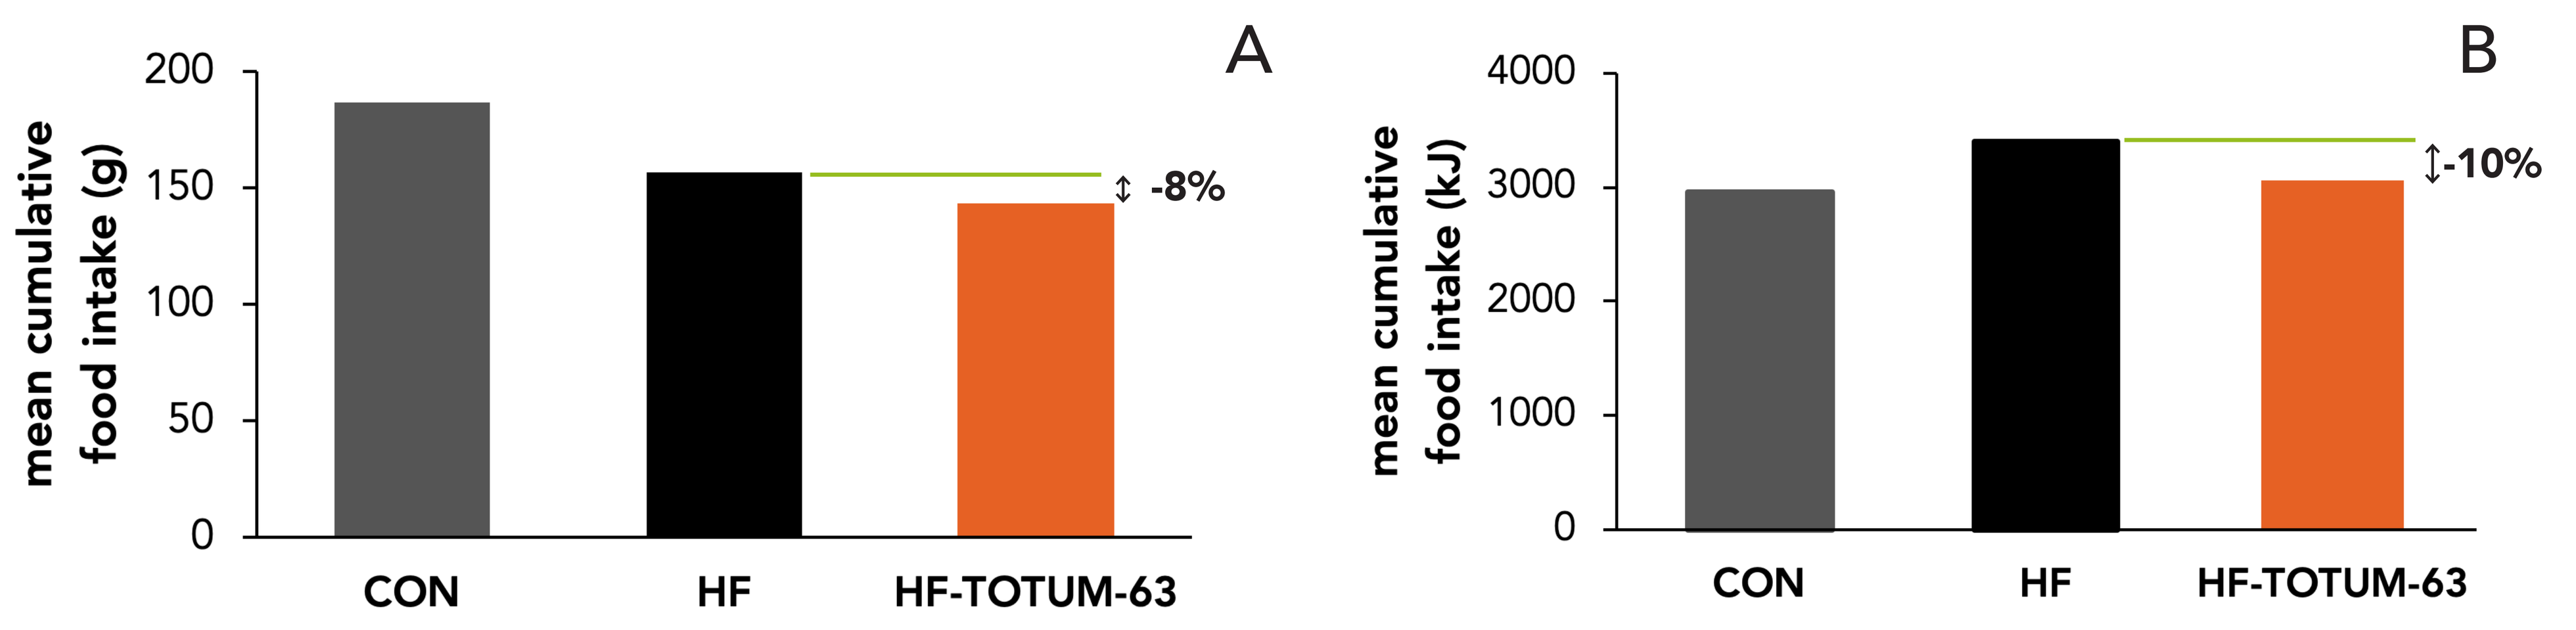
A
mean cumulative food intake (g)
-8%
CON
HF
HF-TOTUM-63
B
mean cumulative food intake (kJ)
-10%
CON
HF
HF-TOTUM-63

Fig. 2 – Impact of TOTUM-63 on cumulative dietary intake in (A) g and in (B) kJ.

Cumulative dietary intake of HF-TOTUM-63 was 8% (in g) and 10% (in kJ) lower compared to that of HF. It was 23% lower (in g) and 3% higher (in kJ) compared to CON.

3. Oral glucose tolerance test

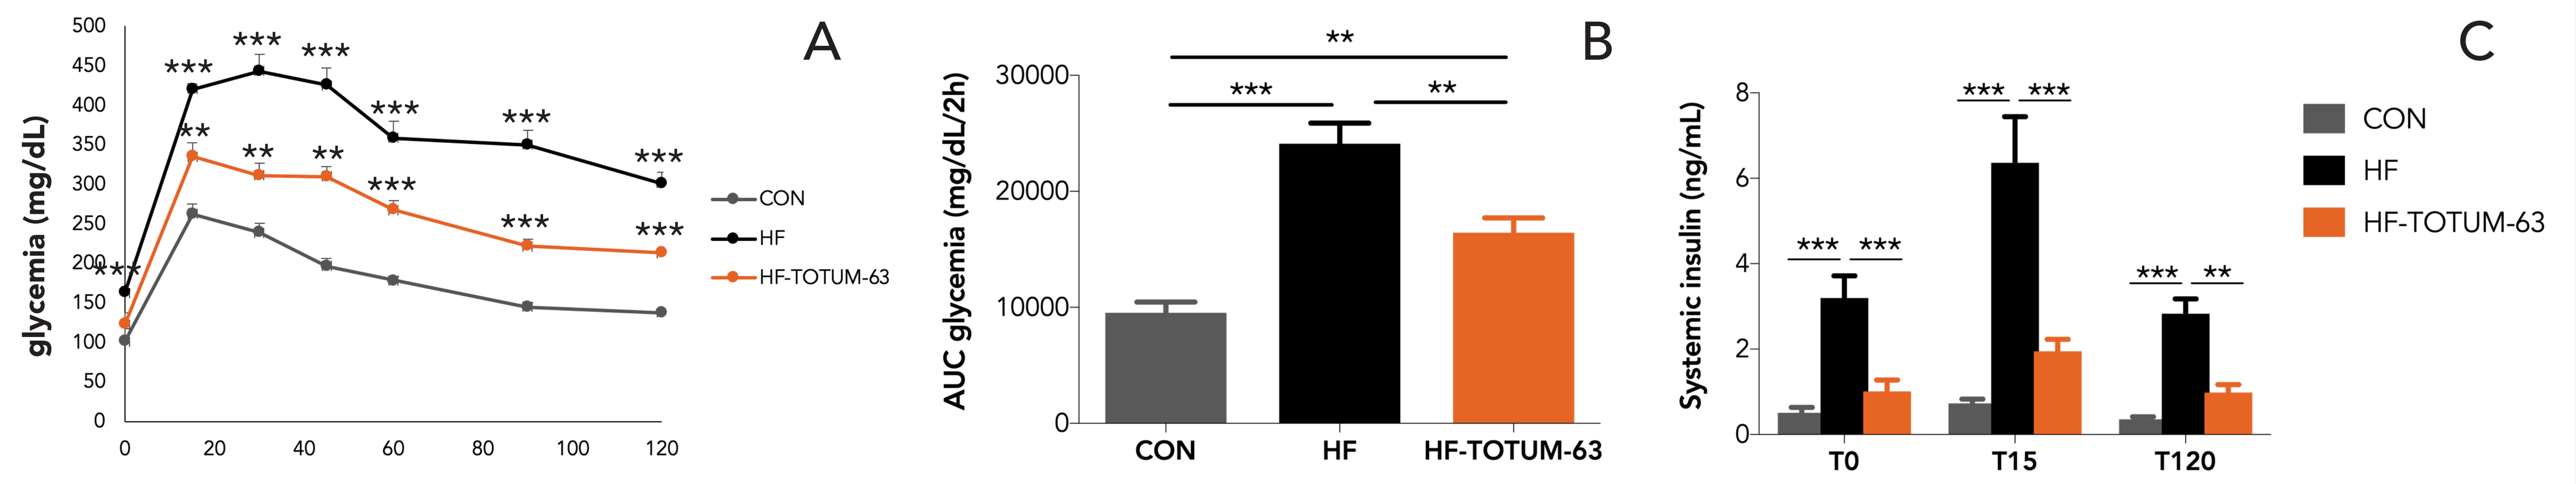
A
glycemia (mg/dL)
CON
HF
HF-TOTUM-63
B
AUC glycemia (mg/dL/2h)
CON
HF
HF-TOTUM-63
C
Systemic insulin (ng/mL)
T0
T15
T120
CON
HF
HF-TOTUM-63

Fig. 3 – Impact of TOTUM-63 on (A) the glycemic response, (B) area under the curve (AUC) of the glycemia and (C) insulin response to the glucose challenge.

**** : p< 0.01;*** : p< 0.001**

OGTT was improved by 31% in HF-TOTUM-63 compared to HF. Insulin response to OGTT was fully restored to CON values.

4. Intraperitoneal pyruvate tolerance test

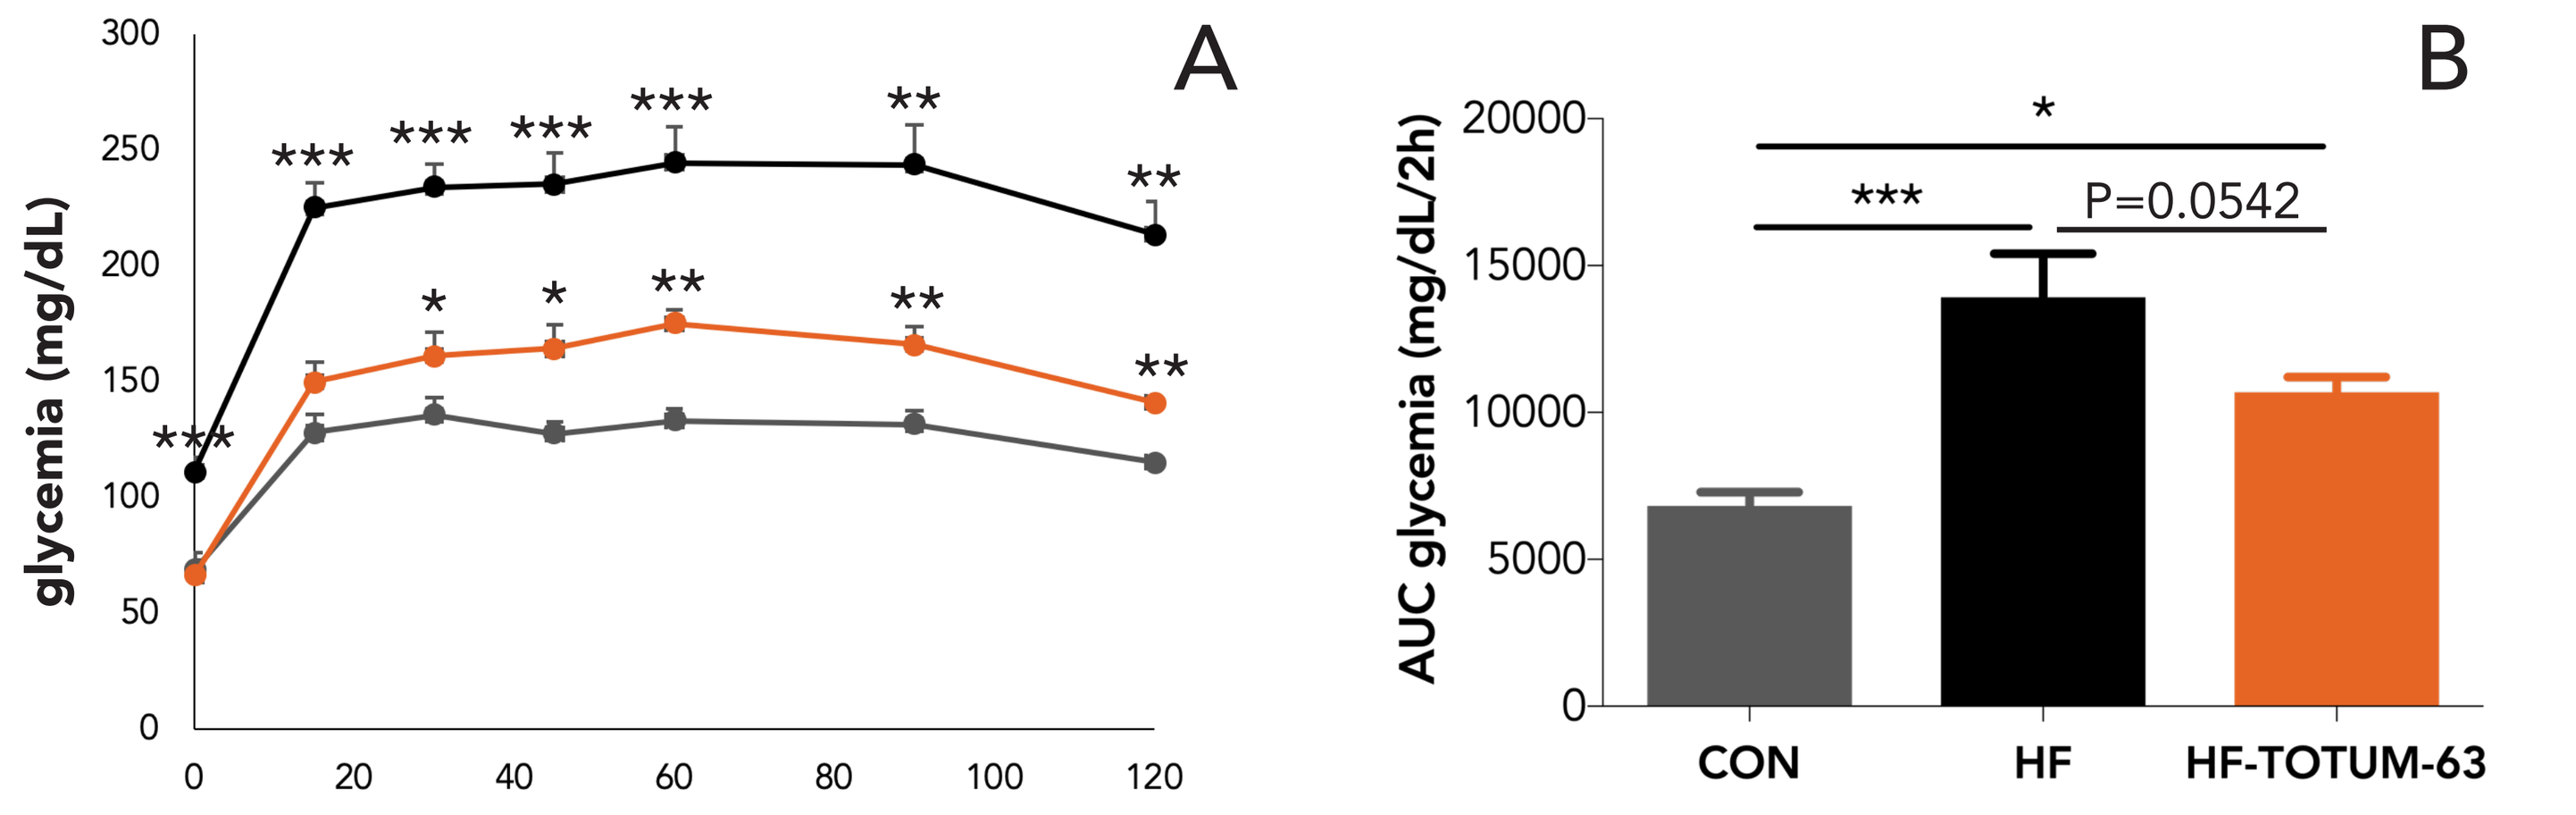
A
glycemia (mg/dL)
B
AUC glycemia (mg/dL/2h)
P=0.0542
CON
HF
HF-TOTUM-63

Fig. 4 – Impact of TOTUM-63 on (A) the glycemic response and (B) area under the curve (AUC) of the glycemia to the pyruvate challenge. * : p< 0.05; ** : p< 0.01; * : p< 0.001**

ipPTT was improved by 23% in HF-TOTUM-63 compared to HF.

5. Portal hormonal responses to glucose stimulation

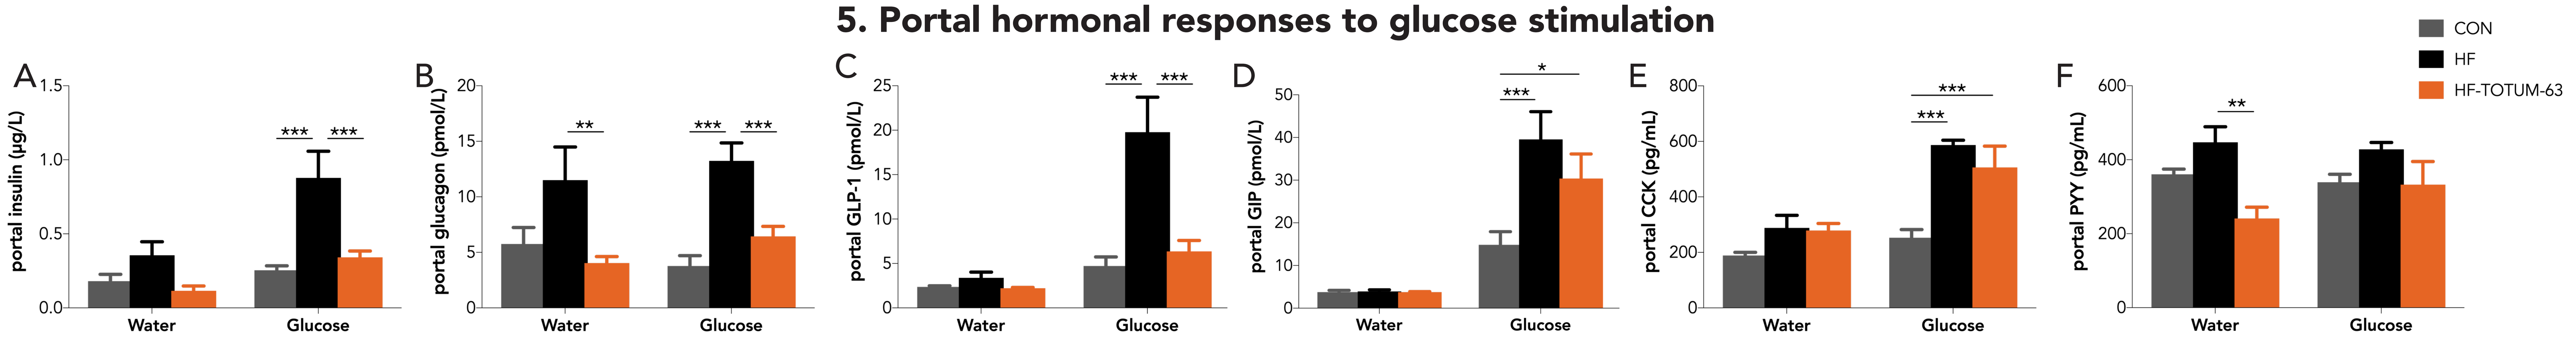
CON
HF
HF-TOTUM-63
A
portal insulin (µg/L)
B
portal glucagon (pmol/L)
C
portal GLP-1 (pmol/L)
D
portal GIP (pmol/L)
E
portal CCK (pg/mL)
F
portal PYY (pg/mL)
Water
Glucose

Fig. 5 – Impact of TOTUM-63 on the portal vein concentration of (A) insulin, (B) glucagon, (C) GLP-1, (D) GIP, (E) CCK and (F) PYY in response to the glucose challenge vs. water. * : p< 0.05; ** : p< 0.01; * : p< 0.001**

Portal concentrations of insulin, glucagon and GLP-1 in response to the glucose challenge were restored almost to the CON level in HF-TOTUM-63, but that of GIP and CCK were still 2 fold higher. Portal concentration of PYY was not significantly altered between treatments after glucose stimulation, it was however significantly decreased by 33 and 46% in unstimulated HF-TOTUM-63-fed animals compared to the CON and HF, respectively.

CarMeN Lyon | Inserm – La science pour la santé – From science to health | INRA Science & Impact | Lyon 1 | VALBIOTIS

EASD 2019
55th Annual Meeting of the European Association for the Study of Diabetes
Barcelona, Spain
16 - 20 September 2019