TOTUM-63 displays beneficial effects on various tissues in animal models of diabetes, via pleiotropic protective actions.

Chavanelle V.[1], Otero Y.F.[1], Sirvent P.[1], Cani P.D.[2], Peltier S[1].

[1]Valbiotis, La Rochelle, France. [2]Louvain Drug Research Institute, Louvain, Belgium.

Background and aims

- About 60 million people are diagnosed with type 2 diabetes (T2D) in Europe with increasing prevalence, mostly due to overweight and obesity (Inzucchi *et al.* 2015).

- Prediabetes precedes T2D and is reversible although 70% of individuals with prediabetes will eventually develop T2D if no intervention is made (Tuso 2014). The pathophysiology of prediabetes is complex and involves impairments in many organs. Additional strategies are needed to manage prediabetes.

- TOTUM-63, a candidate for managing prediabetes, has previously shown beneficial effects on glucose control in rodent models of T2D and in healthy humans, in an open phase I/II clinical trial.

- This study provides new data on 2 different animal models of T2D and prediabetes that show an action of TOTUM-63 on various organs, suggesting a pleiotropic effect of TOTUM-63.

Materials and methods

- All experiments were carried out along the "Principles of laboratory animal care" (NIH Publication no. 85-23, revised 1985) and approved by the local ethics committee (C2EA-02).

- Db/db study: 40 male db/db mice (aged 6 weeks) were fed for 6 weeks either a control diet or the same diet supplemented with 2.7% TOTUM-63.

- High-Fat study: 36 male C57BL6 mice (aged 6 weeks) were randomised into 3 groups. Control (CON) group was fed a control low-fat diet. High Fat (HF) group was fed a HF diet (60% fat). HF-TOTUM-63 group was fed a HF diet supplemented with 2.7% TOTUM-63. Total duration of the HF study was 16 weeks.

- Body weight, fasting glycaemia (Nova Biomedical StatStrip® Xpress) and insulinemia (Alpco Diagnostic ELISA) were monitored monthly (HF study) or weekly (db/db study).

- Body composition was assessed at the end of both studies by EchoMRI™.

- An Oral Glucose Tolerance Test (OGTT) was performed at the end of both studies and consisted of an acute oral glucose gavage of 2.3 g/kg lean mass. Glycaemia and insulinemia were monitored before and after gavage.

- Akt activation was defined as the ratio of pAkt/Akt assessed by western blot in soleus muscle and inguinal fat of HF-fed mice sampled out 10mn after an acute peritoneal injection of insulin (2U/kg).

- Hepatic triglycerides (TG) were assessed using a TG quantification kit (Abcam).

- Caecal content of HF-fed mice was harvested immediately after sacrifice of the animal and frozen into liquid nitrogen. Composition of caecal microbiota was assessed by 16S rDNA taxonomical metasequencing approach (Ondov *et al.*, 2011, Schloss *et al.* 2009). Each taxon (expressed in relative abundance at the phylum, family and genus levels) was analyzed using a one-way (treatment) analysis of variance statistical model (ANOVA) with the MIXED SAS® procedure. For the analysis of the relative abundances of taxa, a Benjamini & Hochberg procedure was applied to control the False Discovery Rate (FDR) due to multiple hypothesis tests on all taxa.

- Values are presented as mean ± SEM. Statistical analysis was conducted on GraphPad Prism 7.0. Normal distribution was tested with a Kolmogorov-Smirnov test and homogeneity of variances by a Bartlett test. Parametric tests (Student t-test for unpaired samples, 1-way ANOVA, repeated-mesure ANOVA and 2-way ANOVA) were applied when both conditions of normality and homogeneity of variances were respected. When these conditions were not respected, non-parametric equivalents (respectively, Mann-Whitney rank comparison test, Krustal-Wallis test or Geisser-Greenhouse corrected 2-way or repeated-mesure ANOVA) were applied.

VALBIOTIS — UCL Université catholique — LDRI Louvain Drug Research Institute

Systemic improvements

In db/db mice

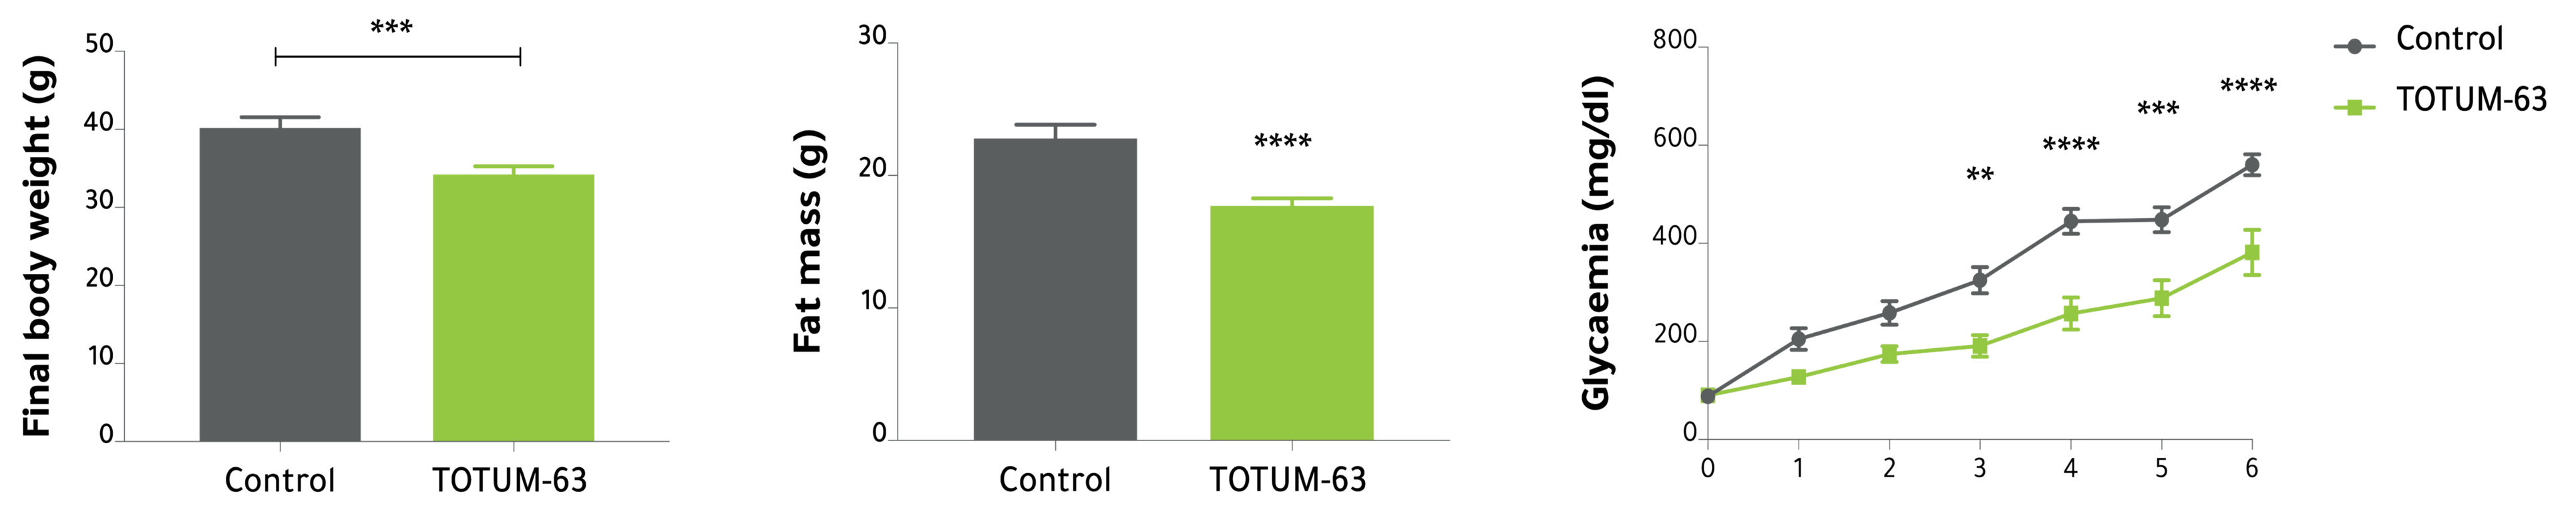
Final body weight (g)
Fat mass (g)
Glycaemia (mg/dl)
Control
TOTUM-63

TOTUM-63 supplemented db/db mice were protected against excessive weight (-15%; p<0.001) and fat mass (-22%, p<0.0001) gain and hyperglycaemia (at week 6, -32%, p<0.0001).

In HF-fed mice

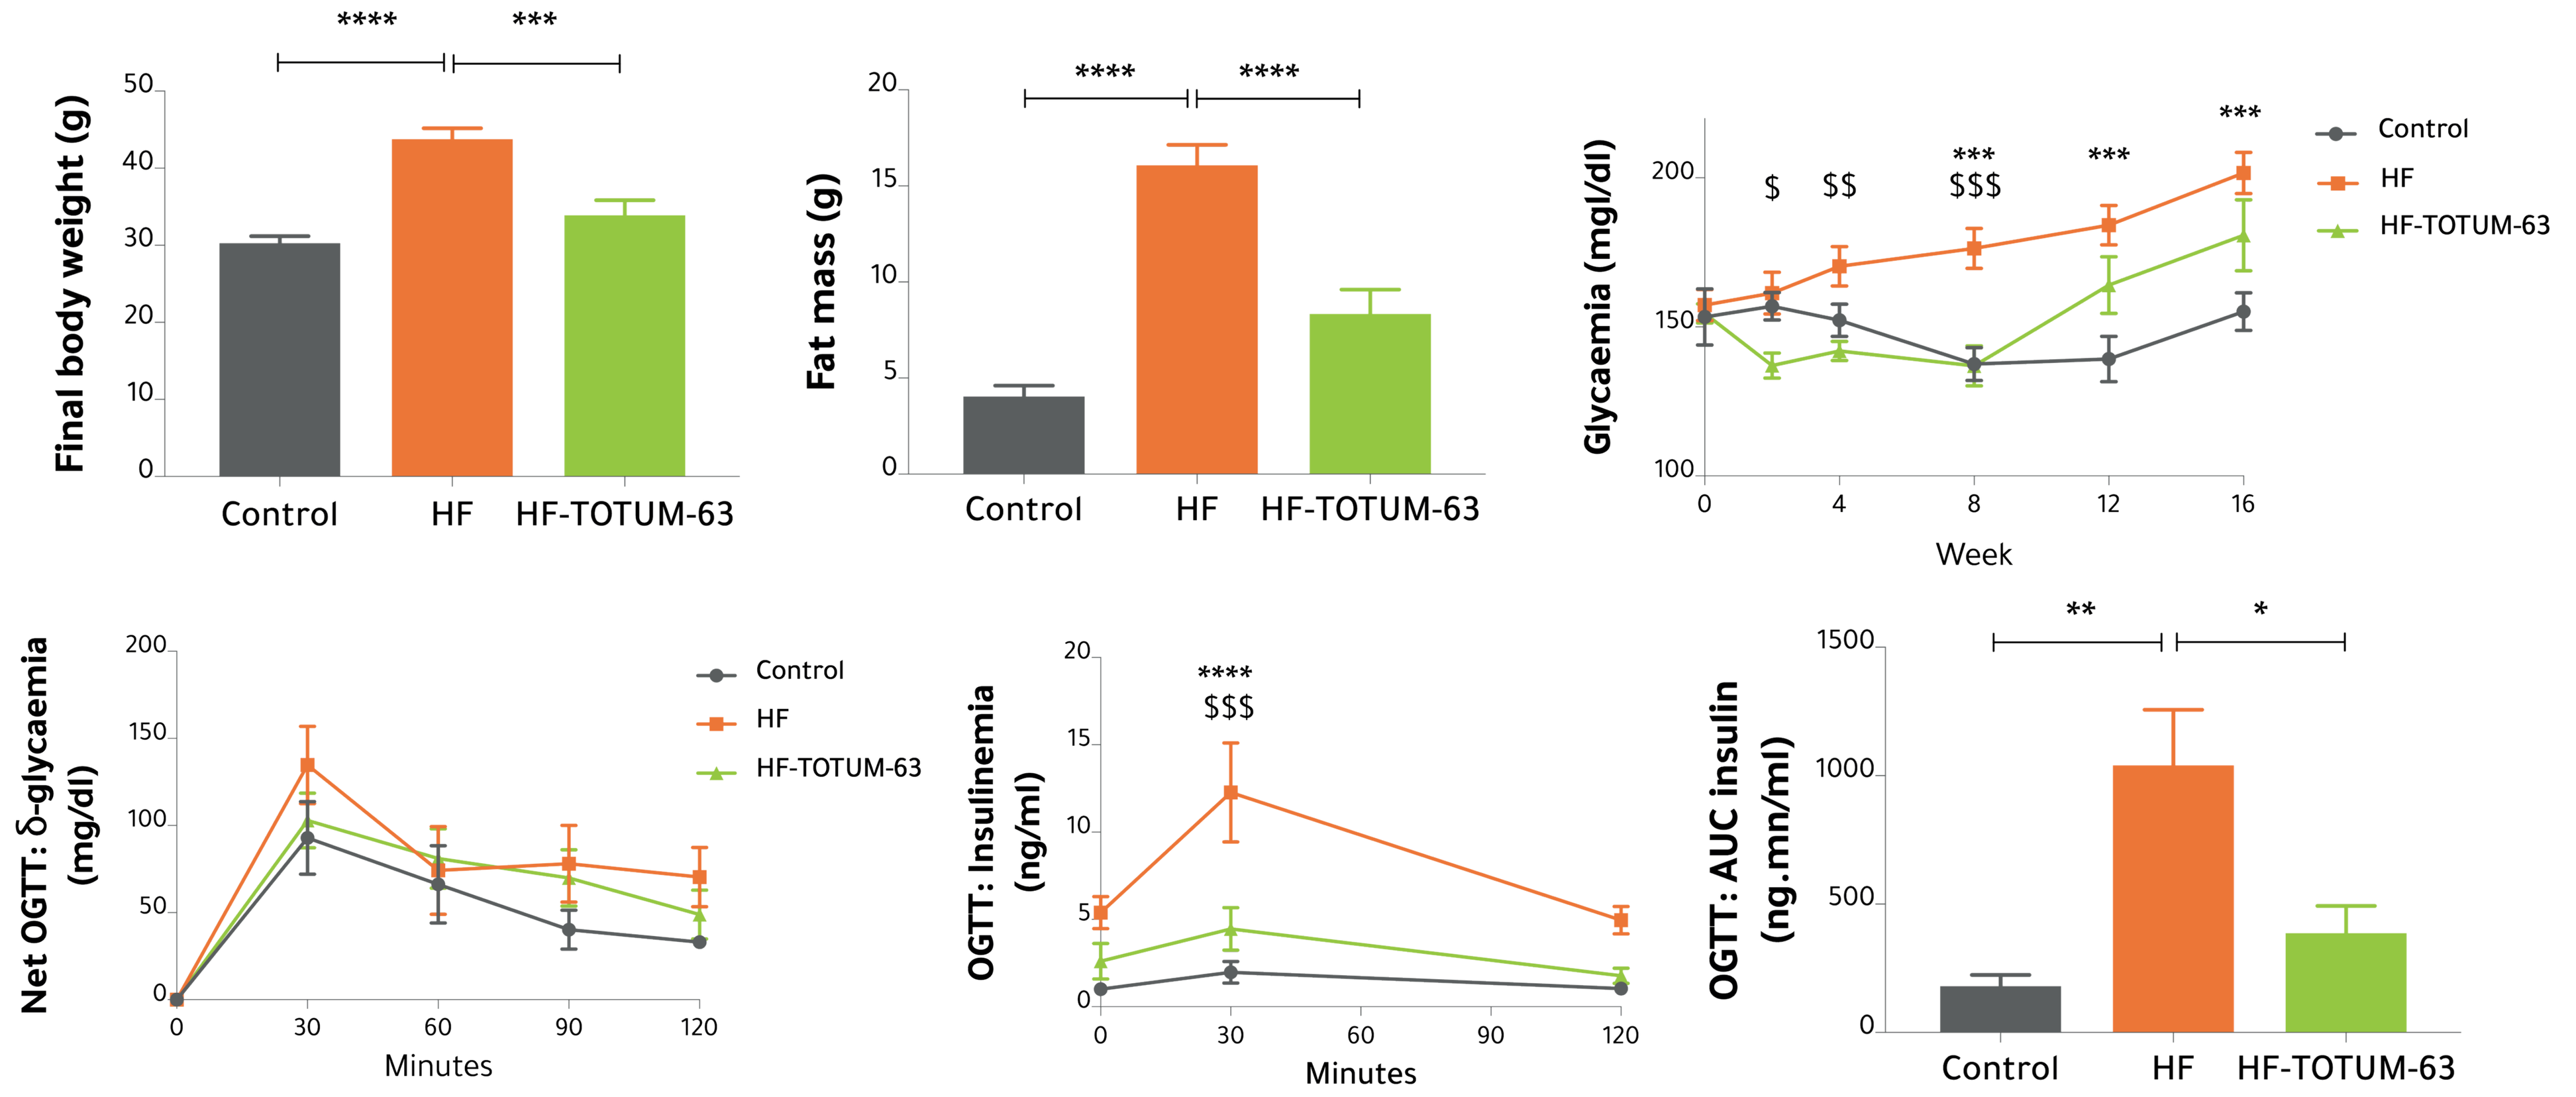
Final body weight (g)
Fat mass (g)
Glycaemia (mgl/dl)
Week
Net OGTT: δ-glycaemia (mg/dl)
OGTT: Insulinemia (ng/ml)
OGTT: AUC insulin (ng.mn/ml)
Minutes
Control
HF
HF-TOTUM-63

TOTUM-63 supplemented HF-fed mice were protected against excessive weight (-23% vs. HF; p<0.001) and fat mass (-48% vs. HF, p<0.0001) gain and hyperglycaemia (at week 8, -22% vs. HF, p<0.001). Insulin peak (at 15mn) and AUC were reduced in HF-TOTUM-63 group (vs. HF, -64% p<0.001 and -63%, p<0.05 respectively) following an oral glucose challenge suggesting improvements on insulin sensitivity. *: different from CON. $: different from HF-TOTUM-63.

Protective action on pancreas

In db/db mice

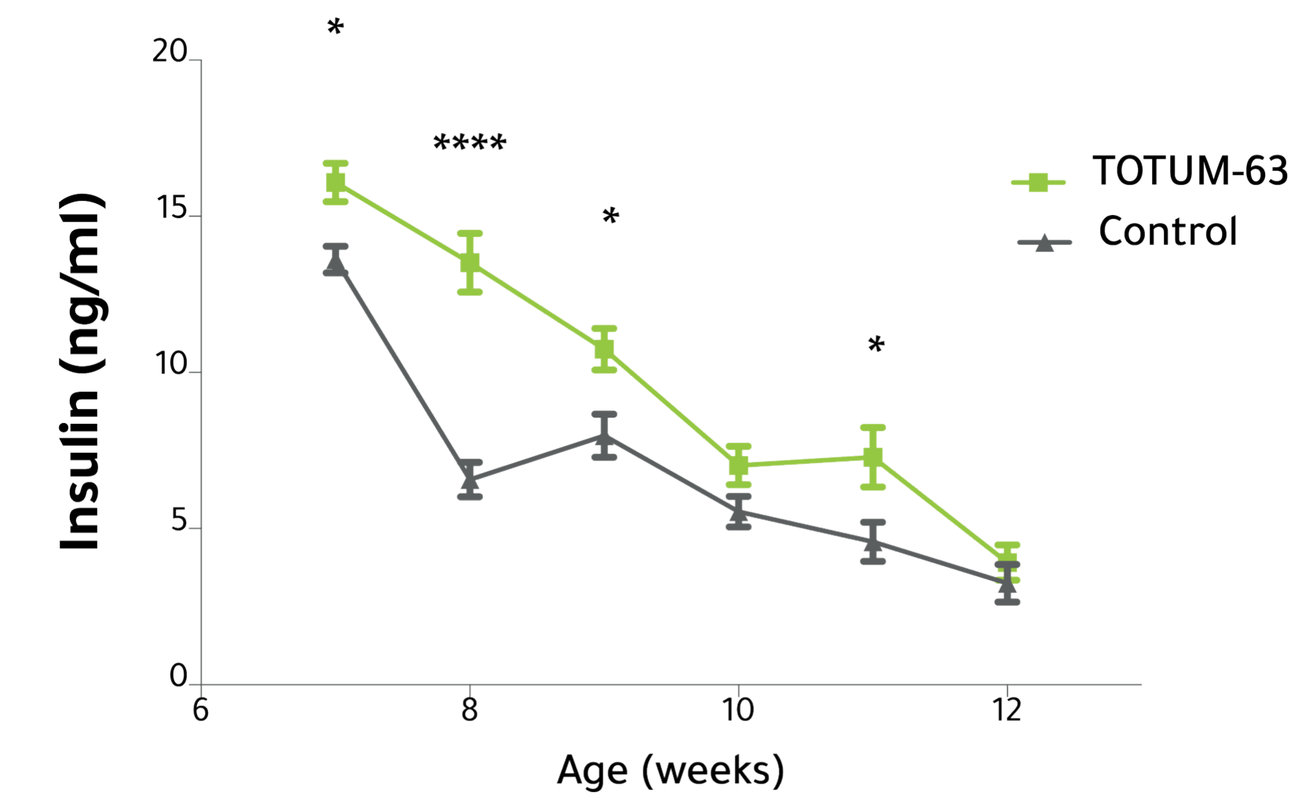
Insulin (ng/ml)
Age (weeks)
TOTUM-63
Control

In db/db mice insulin secretion was severely impaired in CON group after 8 weeks of age. TOTUM-63 may have delayed the defect of insulin secretion, one of the hallmarks of T2D.

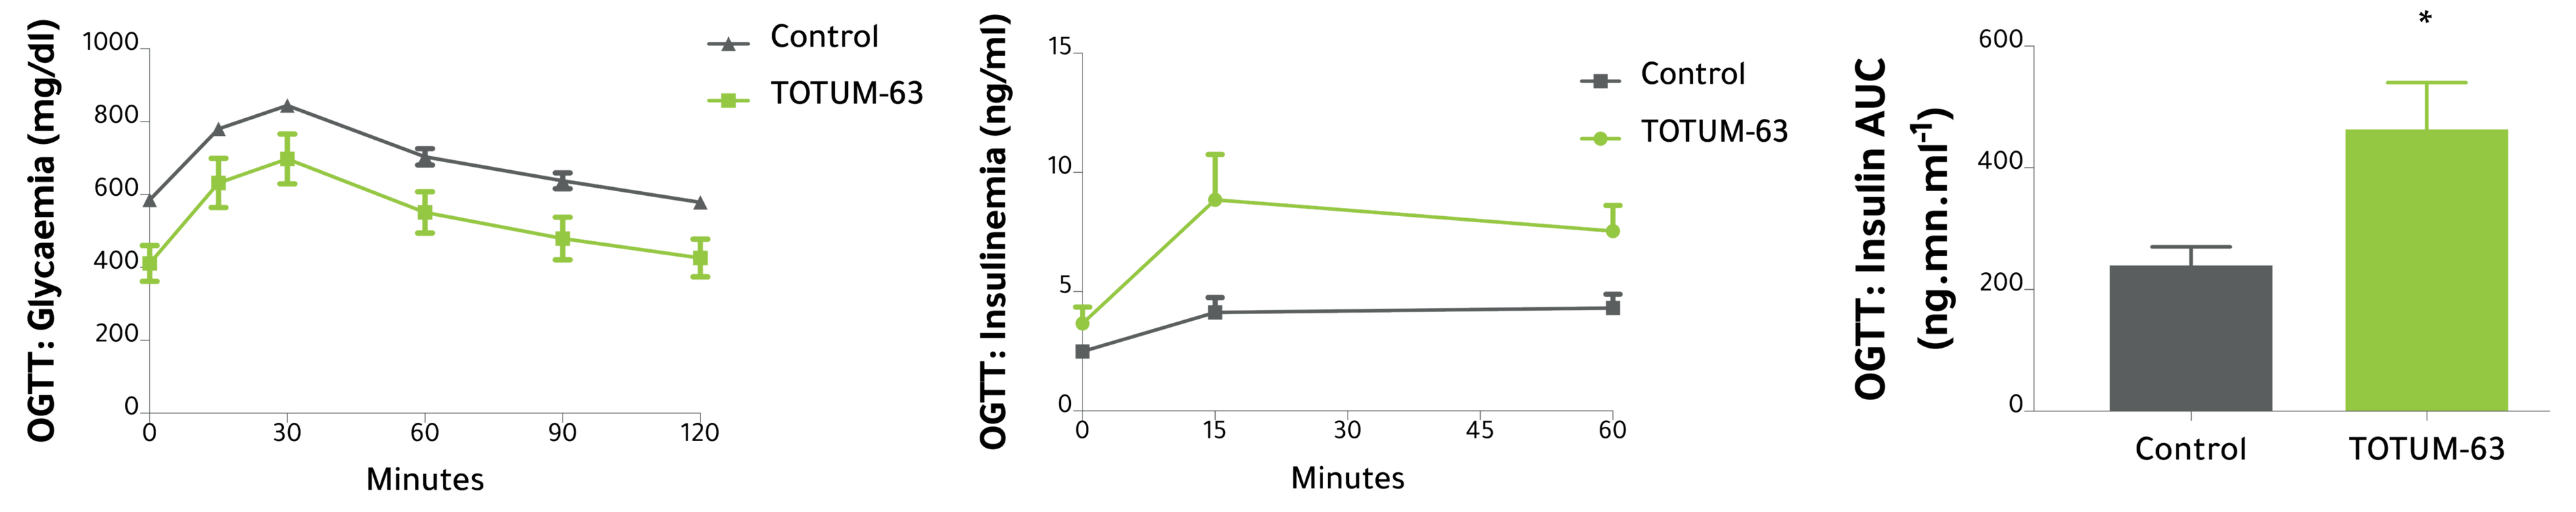
OGTT: Glycaemia (mg/dl)
OGTT: Insulinemia (ng/ml)
OGTT: Insulin AUC (ng.mn.ml-1)
Minutes
Control
TOTUM-63

Similarly, insulin response to an OGTT was blunted in CON, despite hyperglycaemia, but preserved in TOTUM-63. Taken together these data suggest a protective action on the insulin secretion capacity of the pancreas.

Insulin pathway in inguinal fat and soleus muscle

In HF-fed mice

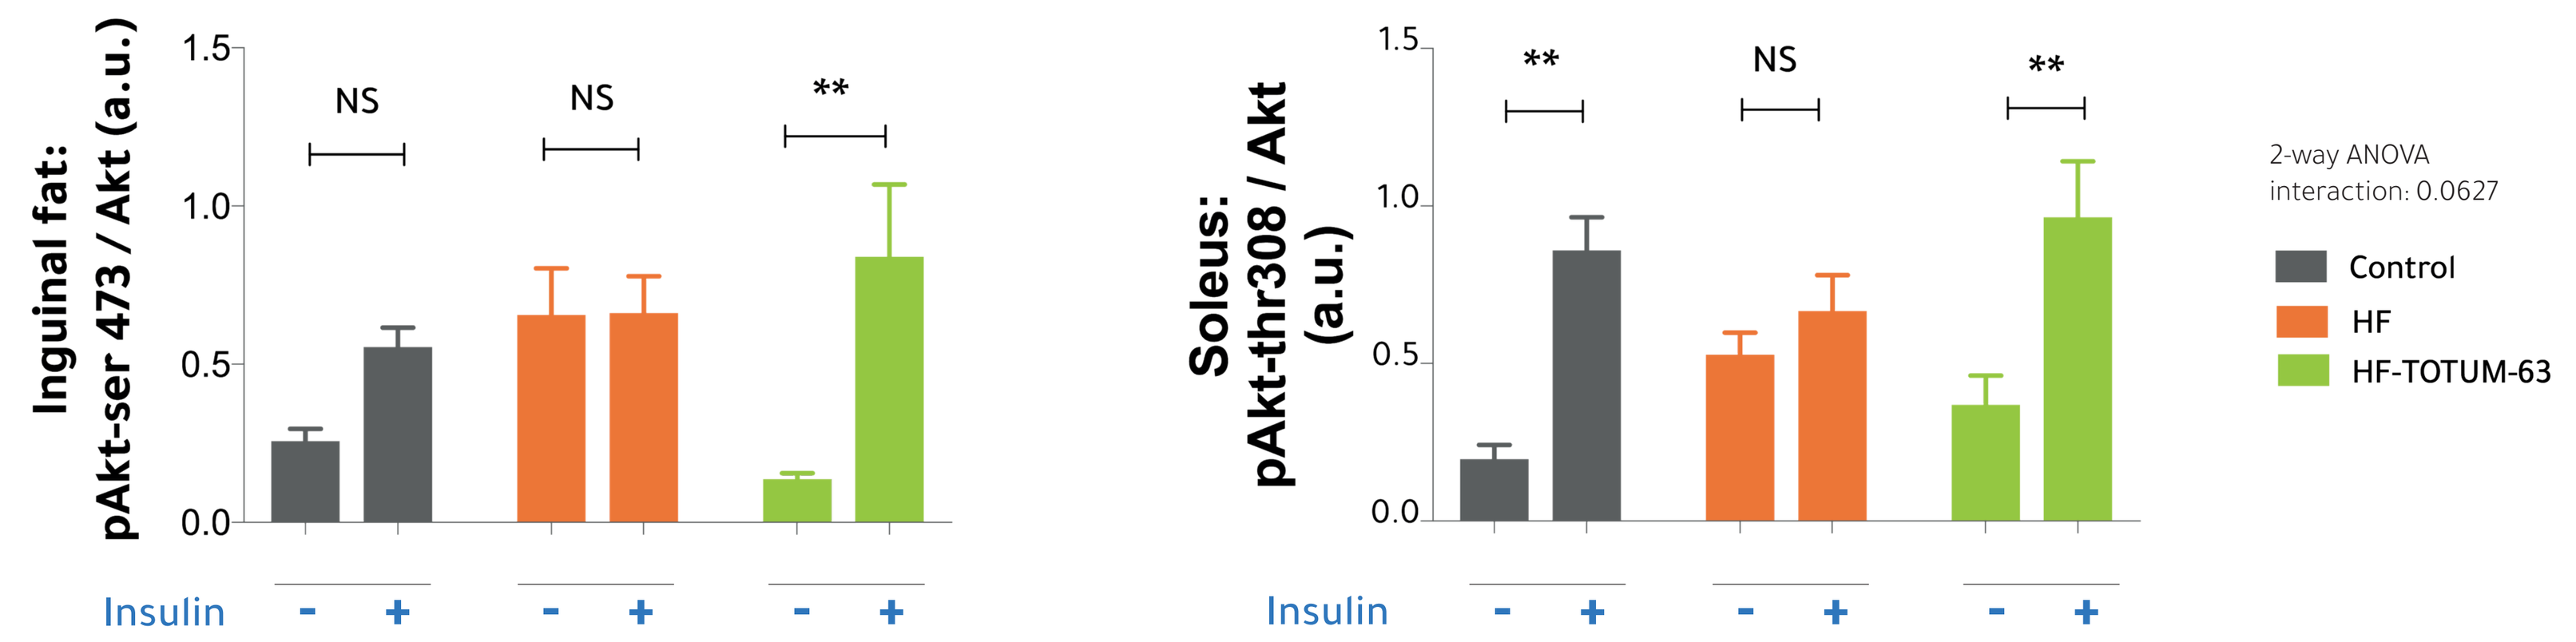
Inguinal fat: pAkt-ser 473 / Akt (a.u.)
Soleus: pAkt-thr308 / Akt (a.u.)
Insulin
2-way ANOVA interaction: 0.0627
Control
HF
HF-TOTUM-63

In the inguinal fat, HF diet impaired the activation of the insulin pathway following an acute injection of insulin. Interestingly, TOTUM-63 restored the activation of Akt in this tissue. A similar trend was observed in soleus muscle, although the interaction (2-way ANOVA) didn't reach statistical significance (p=0.0627).

Liver steatosis

In HF-fed mice

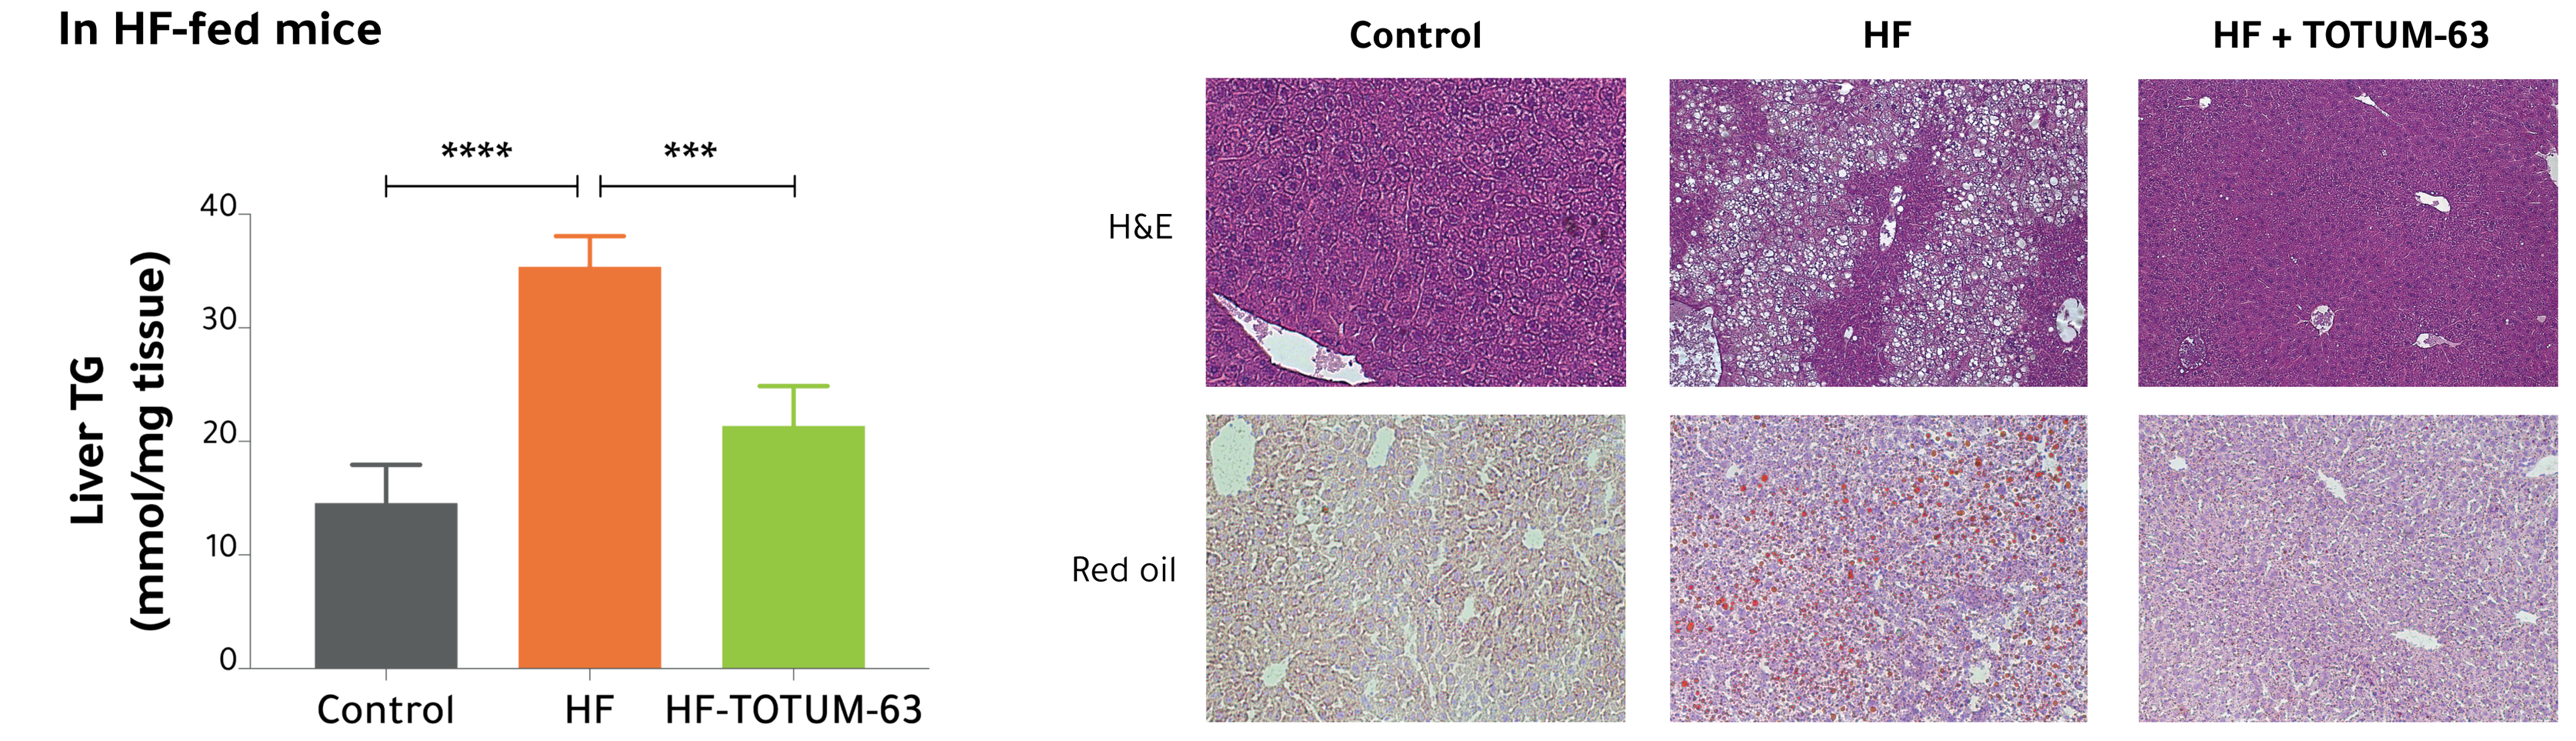
Liver TG (mmol/mg tissue)
Control
HF
HF-TOTUM-63
HF + TOTUM-63
H&E
Red oil

Hepatic triglyceride content, a powerful inducer of liver insulin resistance, was reduced in HF-TOTUM-63 group, compared to HF (-40%, p<0.001). H&E- and Red Oil-stained sections also confirmed the reduced steatosis in TOTUM-63 supplemented animals.

HF-diet induced microbiota dysbiosis

In HF-fed mice

At the genus level, significant differences between HF and HF-Totum63 were found regarding the relative abundance of 10 bacteria. Interestingly, the relative abundance of Parasutterella and Barnesiella, two bacteria possibly linked with improvements in gut disturbances associated with a high fat diet (Zhang et al. 2012, Louis et al. 2016, Bashiardes et al. 2016), was increased in HF-TOTUM-63 group compared to HF (1.55% vs. 0.33%, p<0.001 and 9.36% vs. 3.12%, p<0.001, respectively). These results suggest a preventive effect of TOTUM63 on HF diet-induced gut microbiota dysbiosis.

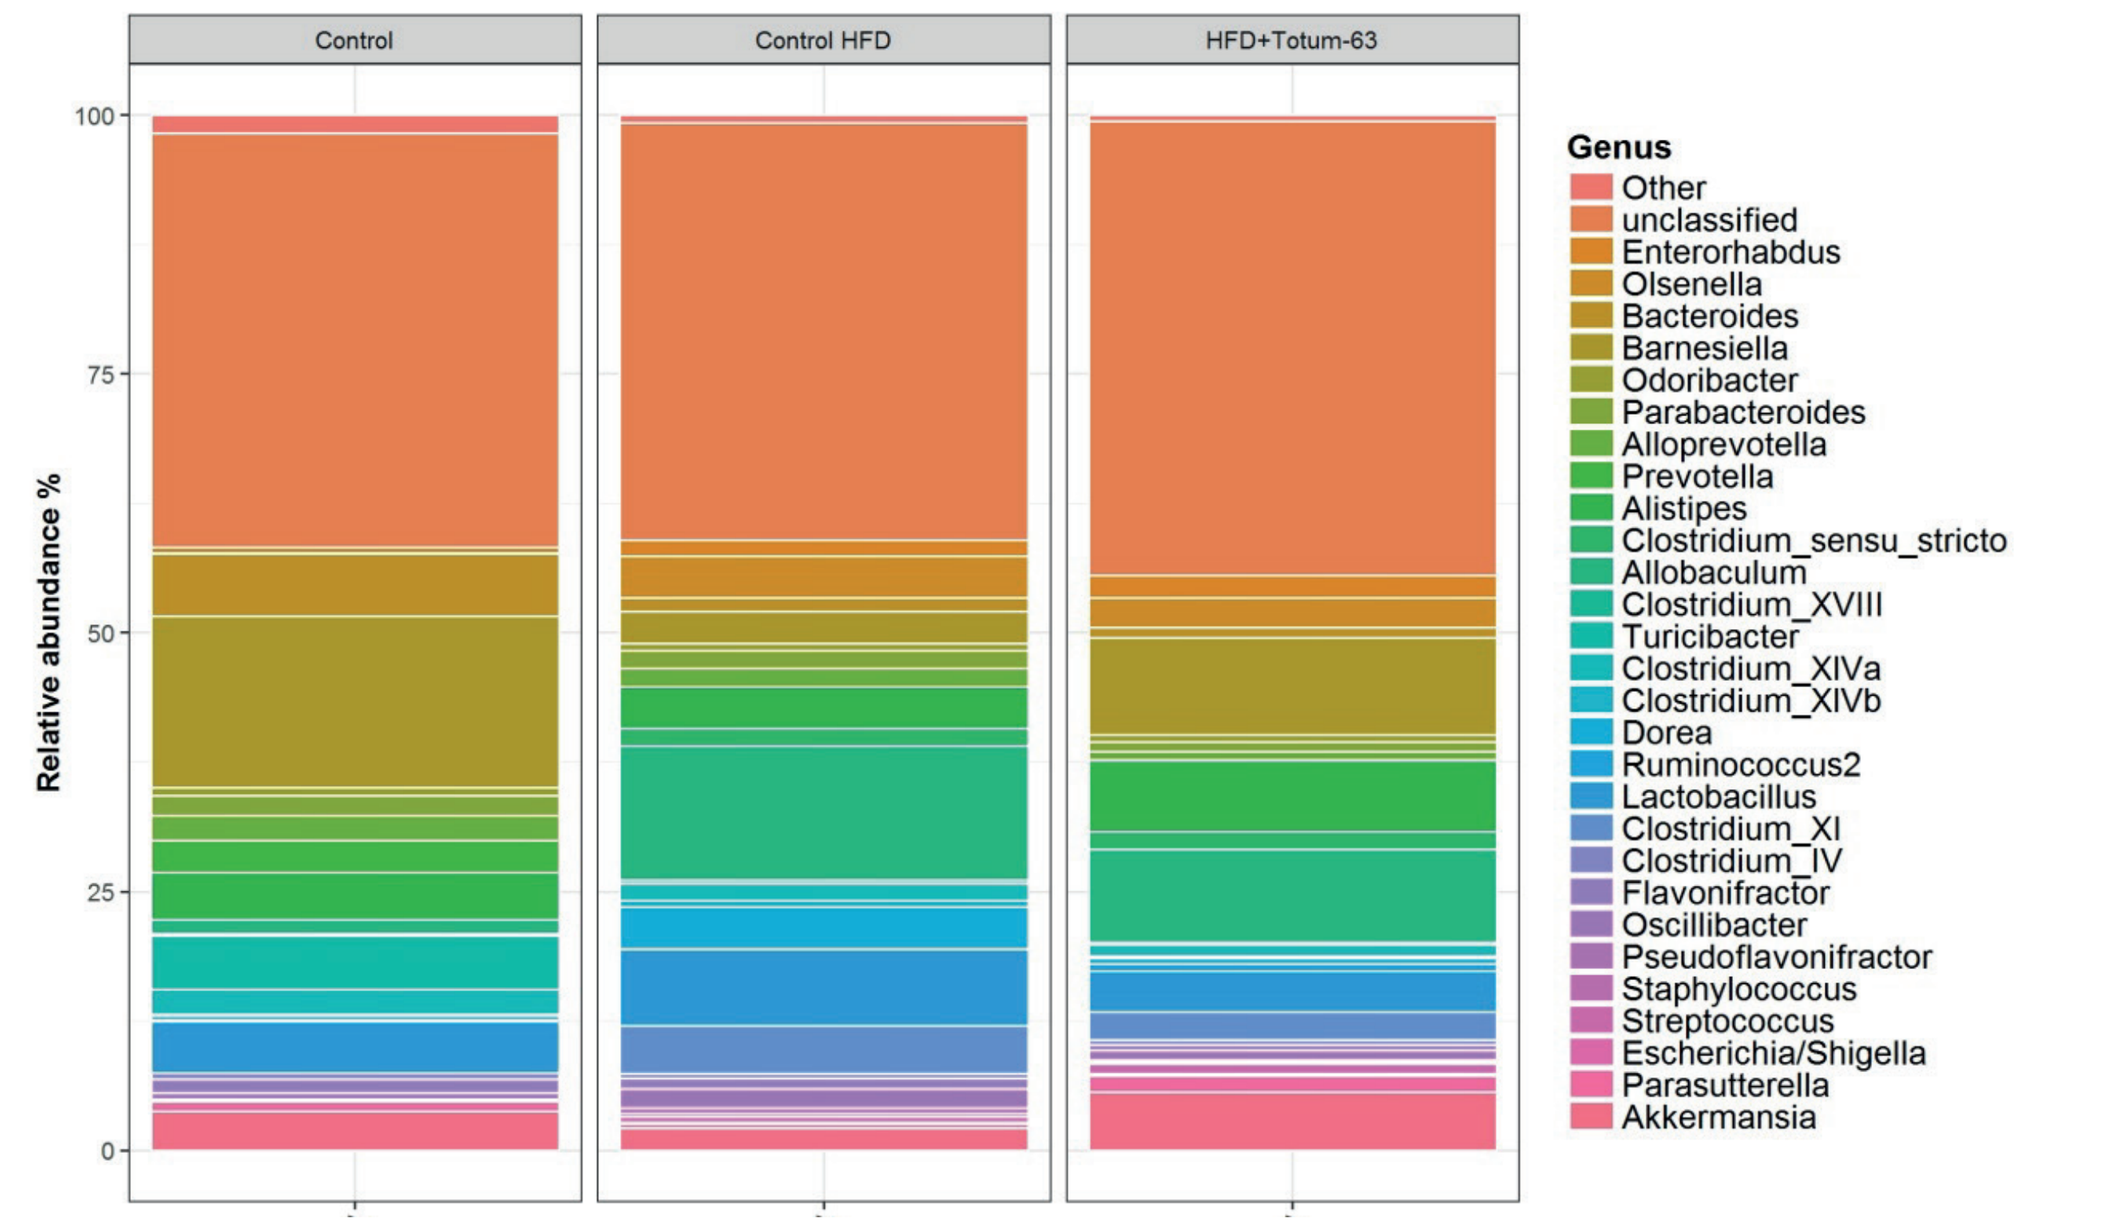
Control
Control HFD
HFD+Totum-63
Relative abundance %
Genus
Other
unclassified
Enterorhabdus
Olsenella
Bacteroides
Barnesiella
Odoribacter
Parabacteroides
Alloprevotella
Prevotella
Alistipes
Clostridium_sensu_stricto
Allobaculum
Clostridium_XVIII
Turicibacter
Clostridium_XIVa
Clostridium_XIVb
Dorea
Ruminococcus2
Lactobacillus
Clostridium_XI
Clostridium_IV
Flavonifractor
Oscillibacter
Pseudoflavonifractor
Staphylococcus
Streptococcus
Escherichia/Shigella
Parasutterella
Akkermansia

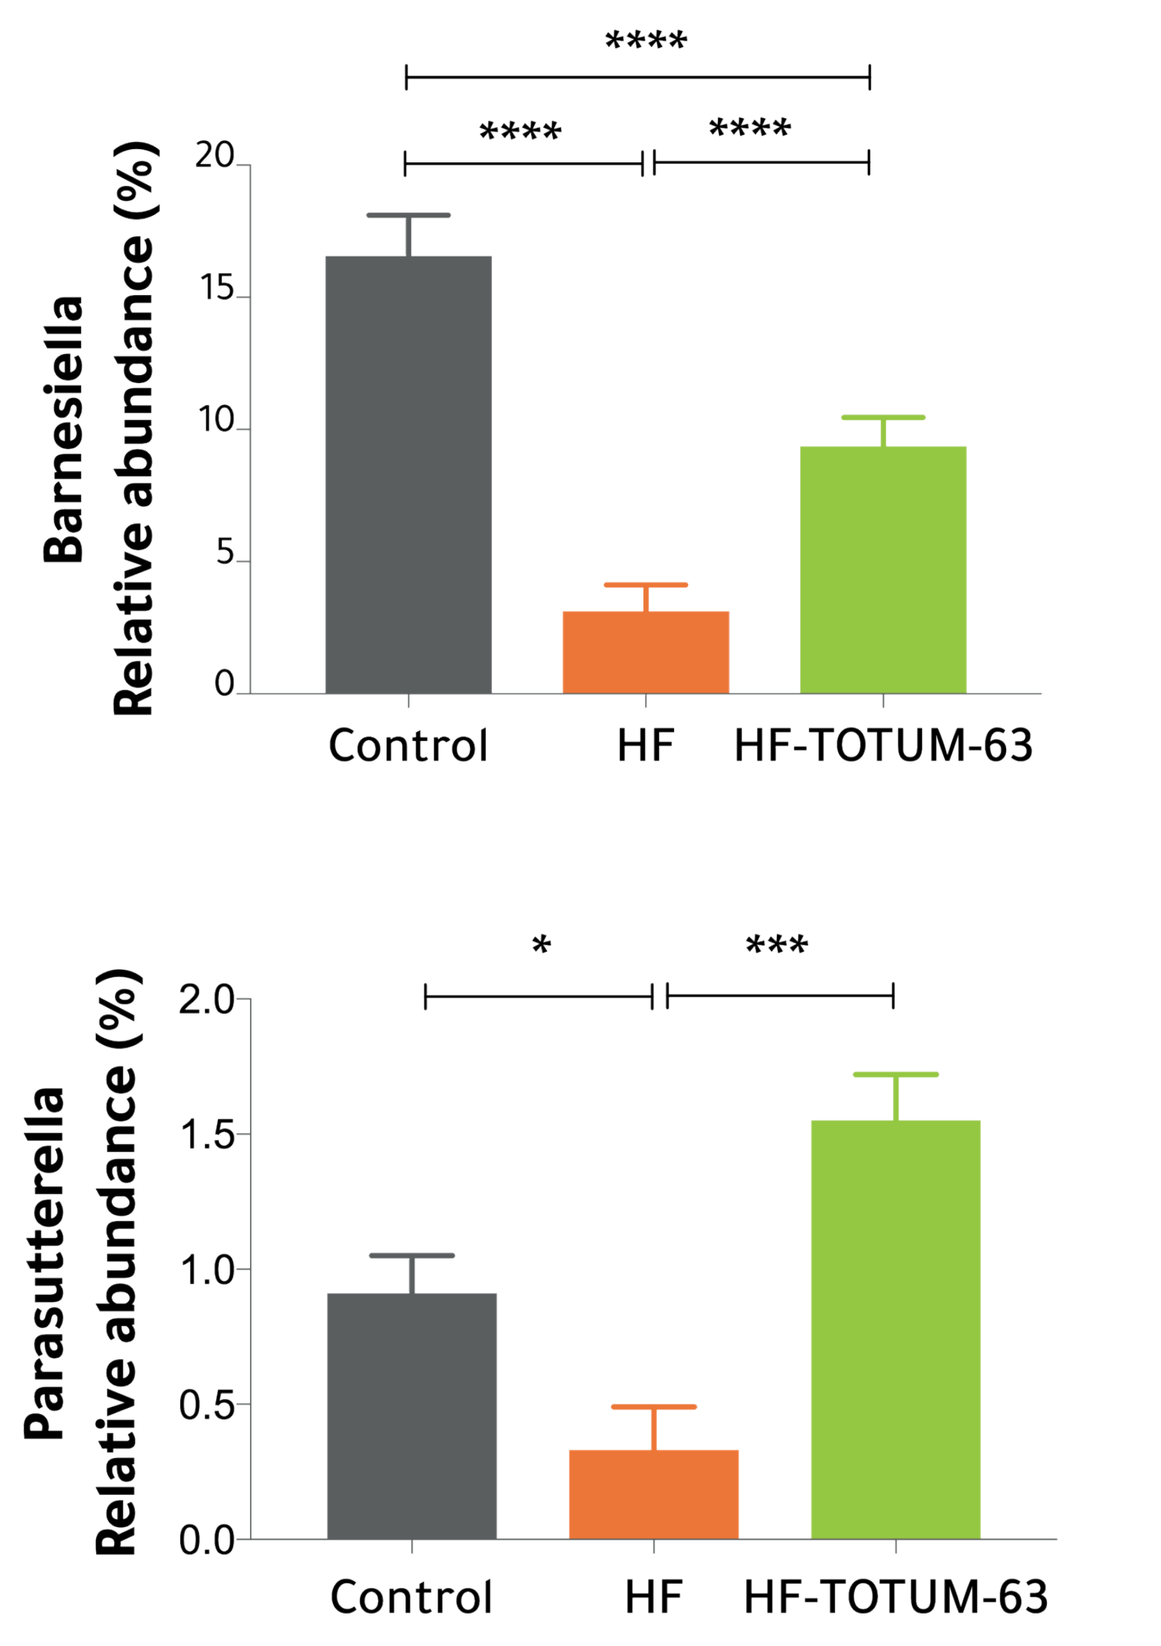
Barnesiella Relative abundance (%)
Parasutterella Relative abundance (%)
Control
HF
HF-TOTUM-63

CONCLUSION

- **TOTUM-63** improved glucose metabolism in 2 mouse models of prediabetes and T2D via actions on various tissues.

- Further experiments are being conducted to confirm the results obtained and investigate other organs potentially involved in T2D pathophysiology (De Fronzo 2008). The pleiotropic effects of TOTUM-63 make it a promising intervention strategy to prevent T2D development.

- The candidate is currently under investigation in an international Phase II clinical trial in prediabetic subjects.

Inzucchi *et al.* (2015); Tuso, P. (2014). The Permanente Journal 18(3); Ondov, B. D. *et al.* (2011) BMC Bioinformatics Sept 30; Schloss P. D. *et al.* (2009) Appl Environ Microbiol 75(23); Inzucchi, S. E. *et al.* (2015). Diabetologia 58(3). Zhang *et al.* (2012). ISME J 6(10). Louis S. *et al.* (2016) PLoS One 11(2) ; Bashiardes *et al.* (2016) Mol metab 5(9); Defronzo, R.A.(2009).Diabetes 58(4)